The Application of COMBINE Analysis to Generate Target-Specific Scoring Functions

Stefan Henrich[1], Ting Wang[1], Niklas Blomberg[2], Rebecca Wade[1]

[1] EML Research gGmbH, Schloss-Wolfsbrunnenweg 33, 69118 Heidelberg, Germany

[2] DECS Chem Comp Dep, SC 2, AstraZeneca R&D Mölndal, S-43183 Mölndal, Sweden

Introduction

Quantitative structure-activity relationship (QSAR) analysis is an essential method to correlate the properties of a series of molecules with their biological activities and to predict the activities of new compounds. Tailor-made scoring functions can be constructed by using structure-based COMparative BINding Energy (COMBINE) analysis (ref. 1-3). This method provides the possibility to derive 3D QSARs for a set of receptor-ligand complexes whose 3D structures can be modeled. The resultant QSARs can guide modifications of either receptor, e. g. in protein engineering, or ligand, e. g. in drug design.

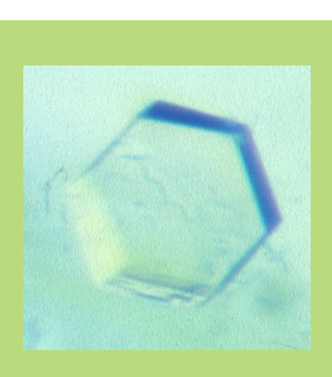

Crystal, NMR or modeled structures of complexes of receptor and ligand

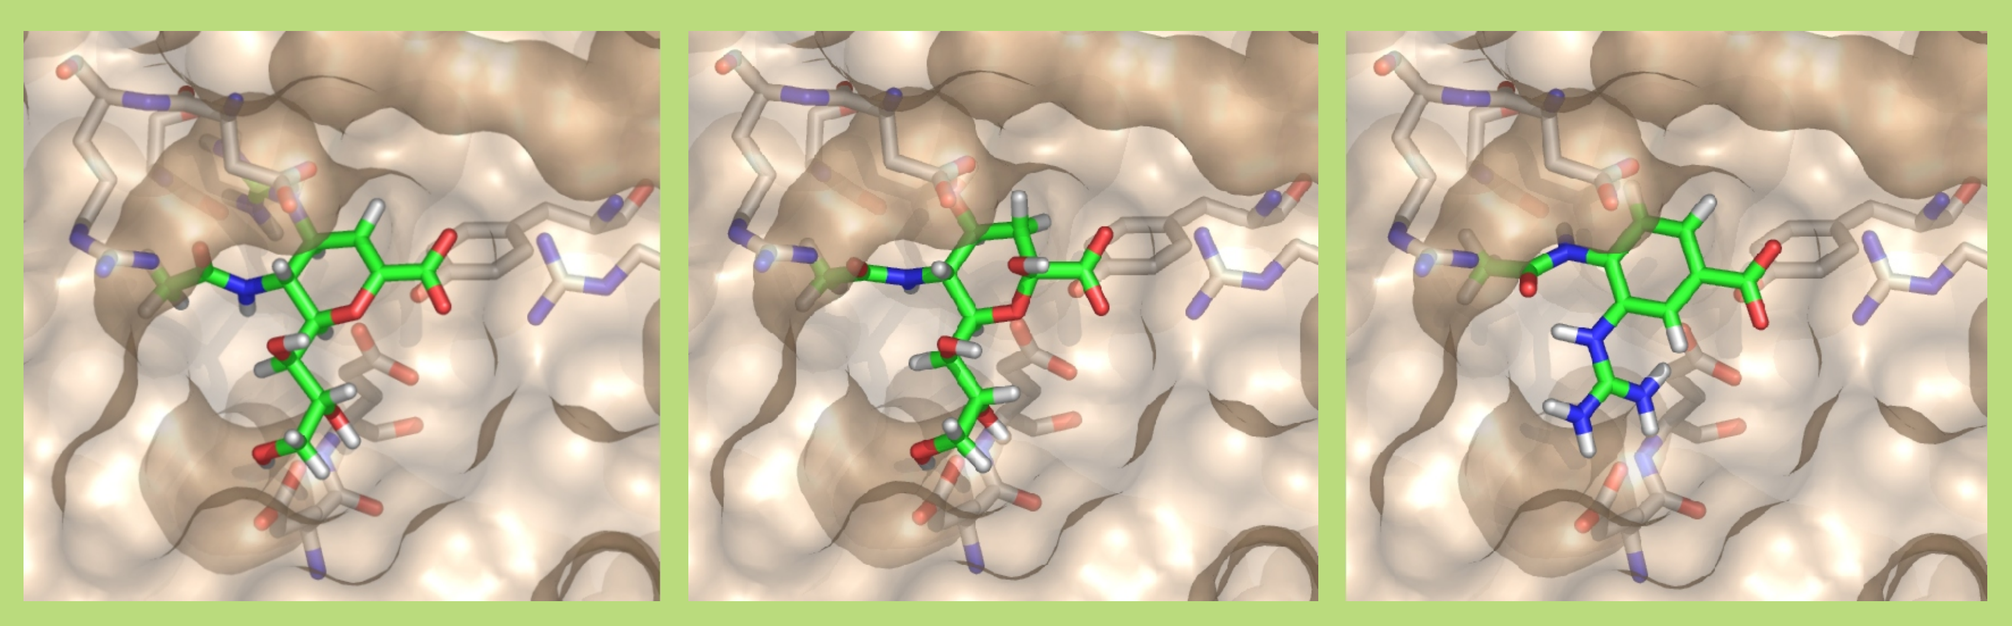

Optimization of hydrogen bond network with WHATIF

Molecular mechanics energy minimization of all models in AMBER8

Calculate ligand-receptor binding energy for each complex with ANAL module of AMBER8
U= intermolecular interaction energy
+ changes in bonded and nonbonded energies of the receptor and the ligands

Partition U of the receptor and the ligand into several components on basis of location in the complex and physicochemical properties

Principle Component Analysis (PCA) in GOLPE4.6

Experimental binding data of protein-ligand-interactions (e.g., K_i values)

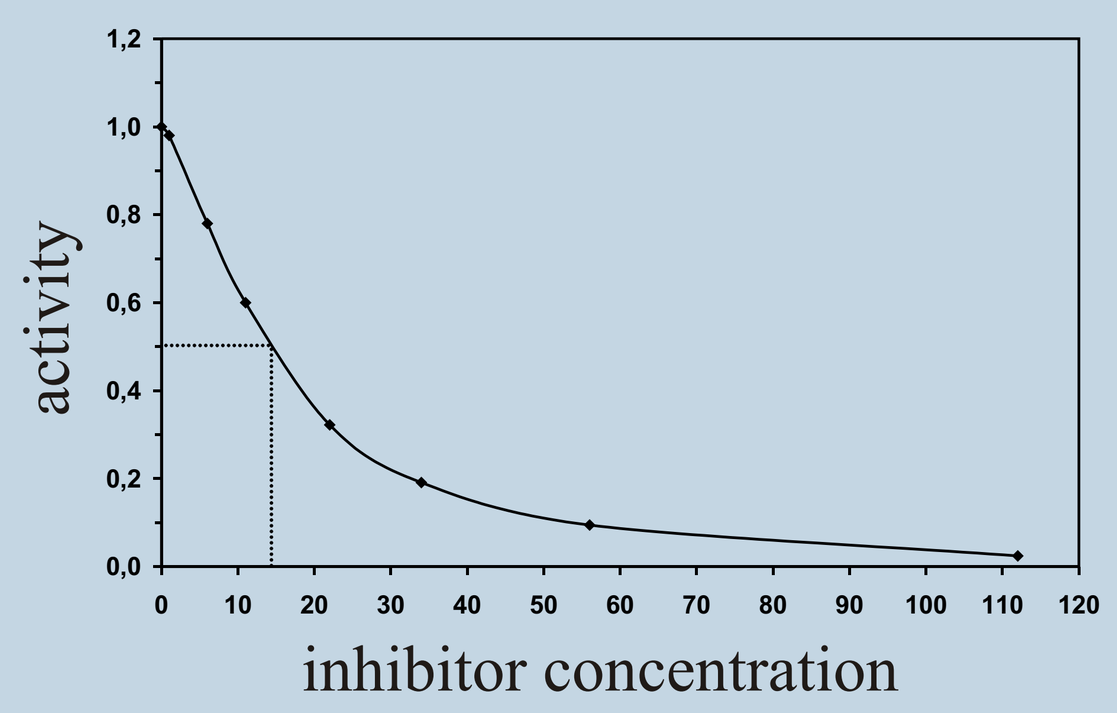
activity
1,2
1,0
0,8
0,6
0,4
0,2
0,0
0
10
20
30
40
50
60
70
80
90
100
110
120
inhibitor concentration

Correlate different components of binding energy U (X variables) with G/activity values (Y variables) by Partial Least Squares (PLS) in GOLPE4.6 for generating target-specific scoring functions. Binding free energy (or biological activities) can be modelled as

$$G = \sum_{i=1}^{n} {}^{vdw}_{i}\, u_i^{vdw} + \sum_{i=1}^{n} {}^{ele}_{i}\, u_i^{ele} + \sum_{i=1}^{n} {}^{desolv}_{i}\, u_i^{desolv} + C$$

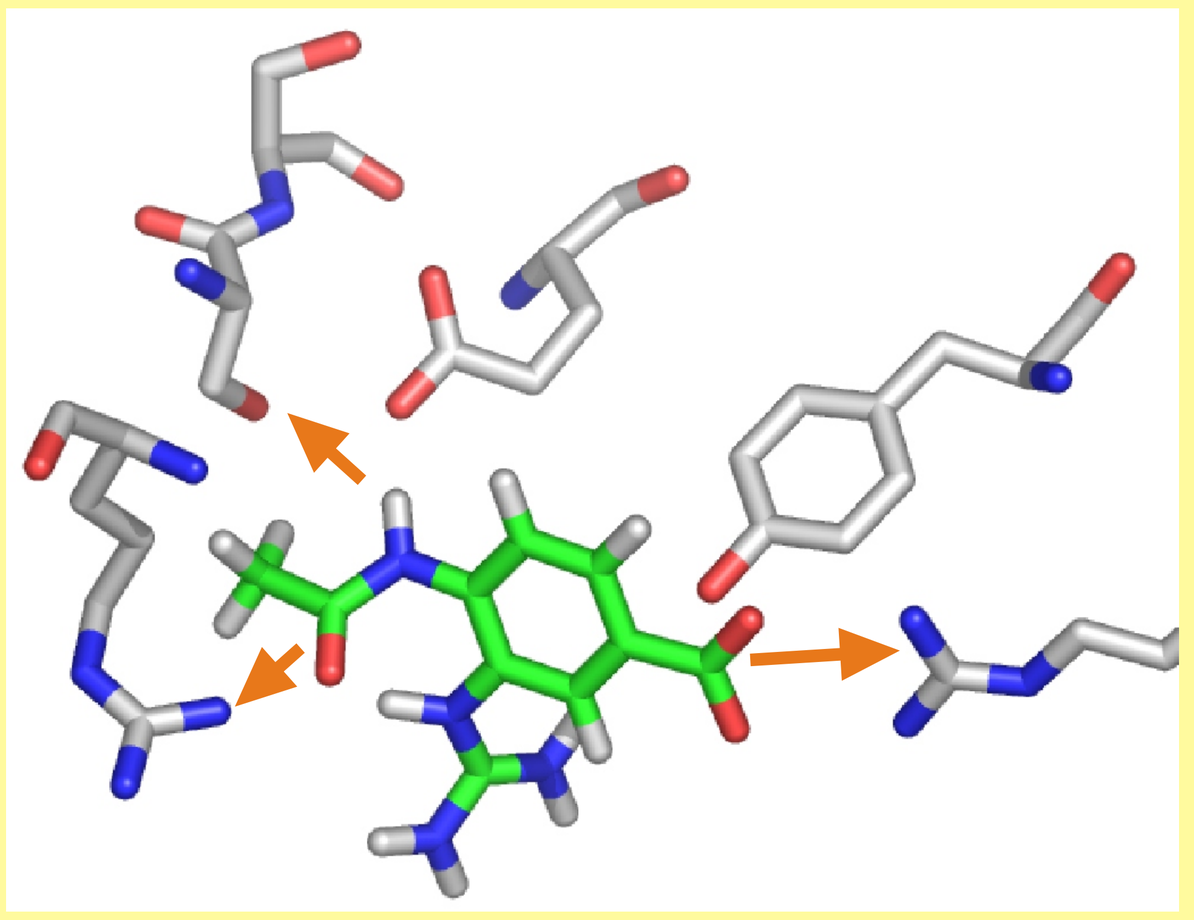

Identify important interactions for determining G/activity

Generated 3D coordinates of small molecule libraries

Flexible docking into target receptors

Molecular mechanics energy minimization of docked ligand and receptor model in AMBER8

Predict activity values for docked compounds

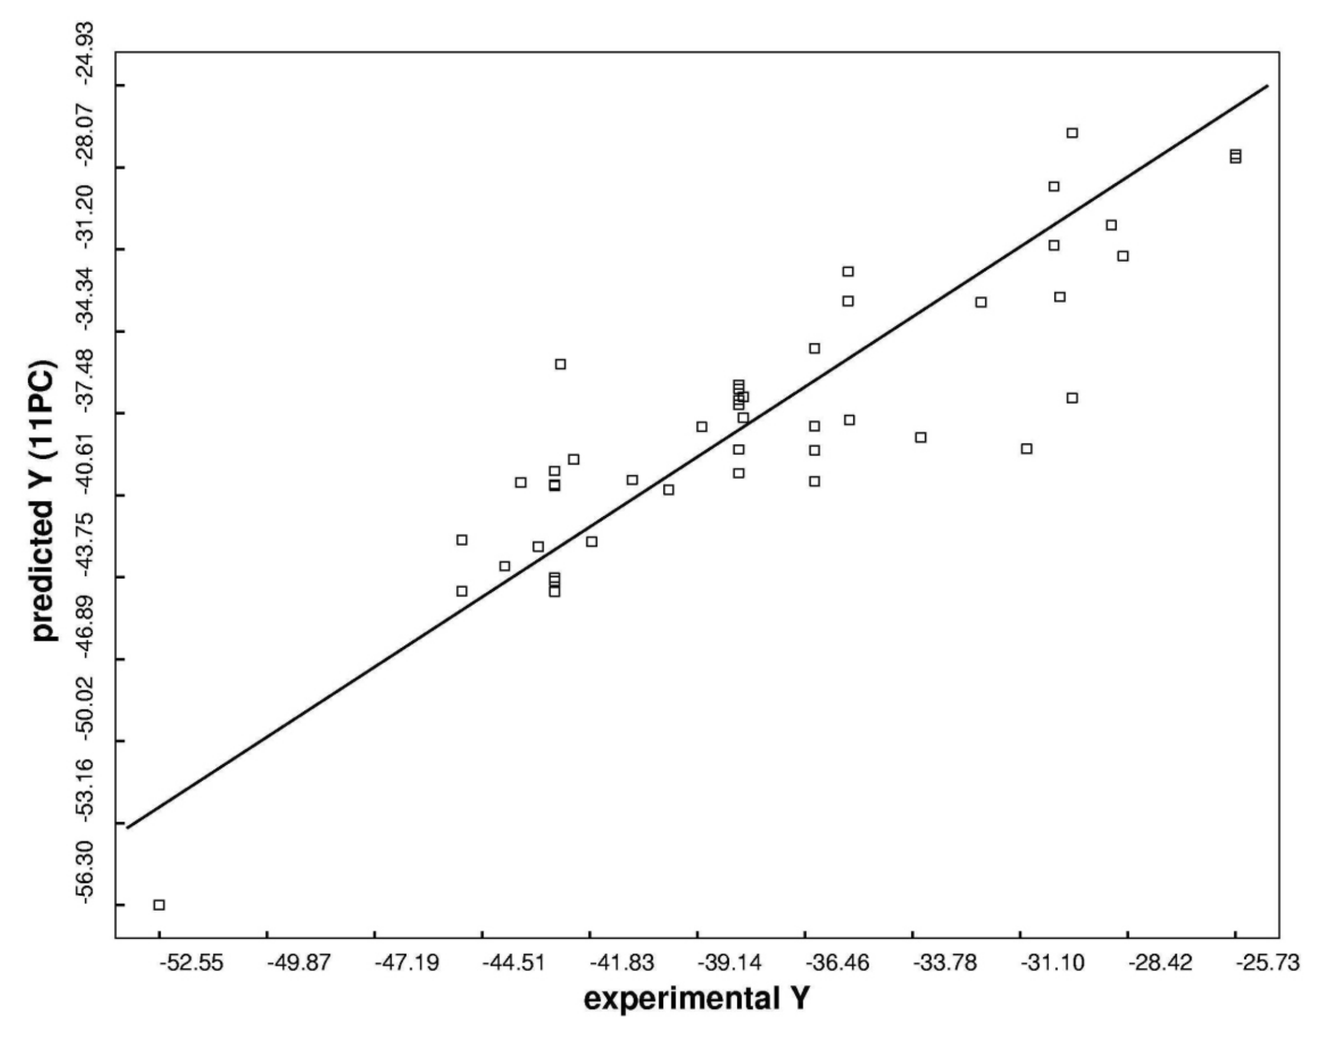
predicted Y (11PC)
-56.30
-53.16
-50.02
-46.89
-43.75
-40.61
-37.48
-34.34
-31.20
-28.07
-24.93
-52.55
-49.87
-47.19
-44.51
-41.83
-39.14
-36.46
-33.78
-31.10
-28.42
-25.73
experimental Y

Method

The principal idea of COMBINE method is the assumption that the binding free energy G is correlated with a subset of weighted interaction energies determined by structures of receptor and ligand. COMBINE analysis starts with an energy minimized model of a receptor-ligand complex that is divided for energy calculations into parts according to their spatial location, normally its amino acid residues. These parts are used together with the ligand (and some important bound water molecules) for calculating electrostatic and van de Waals interaction energies (and desolvation energy terms) between parts of the ligand and of the receptor.

The resultant energy terms of many receptor-ligand complexes are analysed by Principial Component Analysis (PCA) and are correlated to activity values by Partial Least Squares (PLS) coupled with suitable variable selection and data pretreatment. With this correlation, important residues of the target can be pointed out for describing the binding affinity between receptor and ligand.

New ligands which are docked into the active site can be ranked and activity can be predicted (target-specific scoring function).

The Targets

COMBINE analysis has been applied to many different types of receptor-ligand complexes. For example, in the case of influenza neuraminidase (ref. 4), where targets were several subtypes and mutants, the binding affinity of inhibitors could be predicted.

In the present work we apply the COMBINE analysis approach to the problem of predicting the selectivity against different trypsin-like serine proteases of the blood coagulation cascade, because of the large amount of published data as well as their importance in diseases. Important in the development of new selective inhibitors is the high structural similarity between the different proteases (figure 1). With target-specific scoring functions generated by COMBINE analysis, we will specify residues of the different receptors which are relevant for ligand binding. This information can be used for designing new selective ligands.

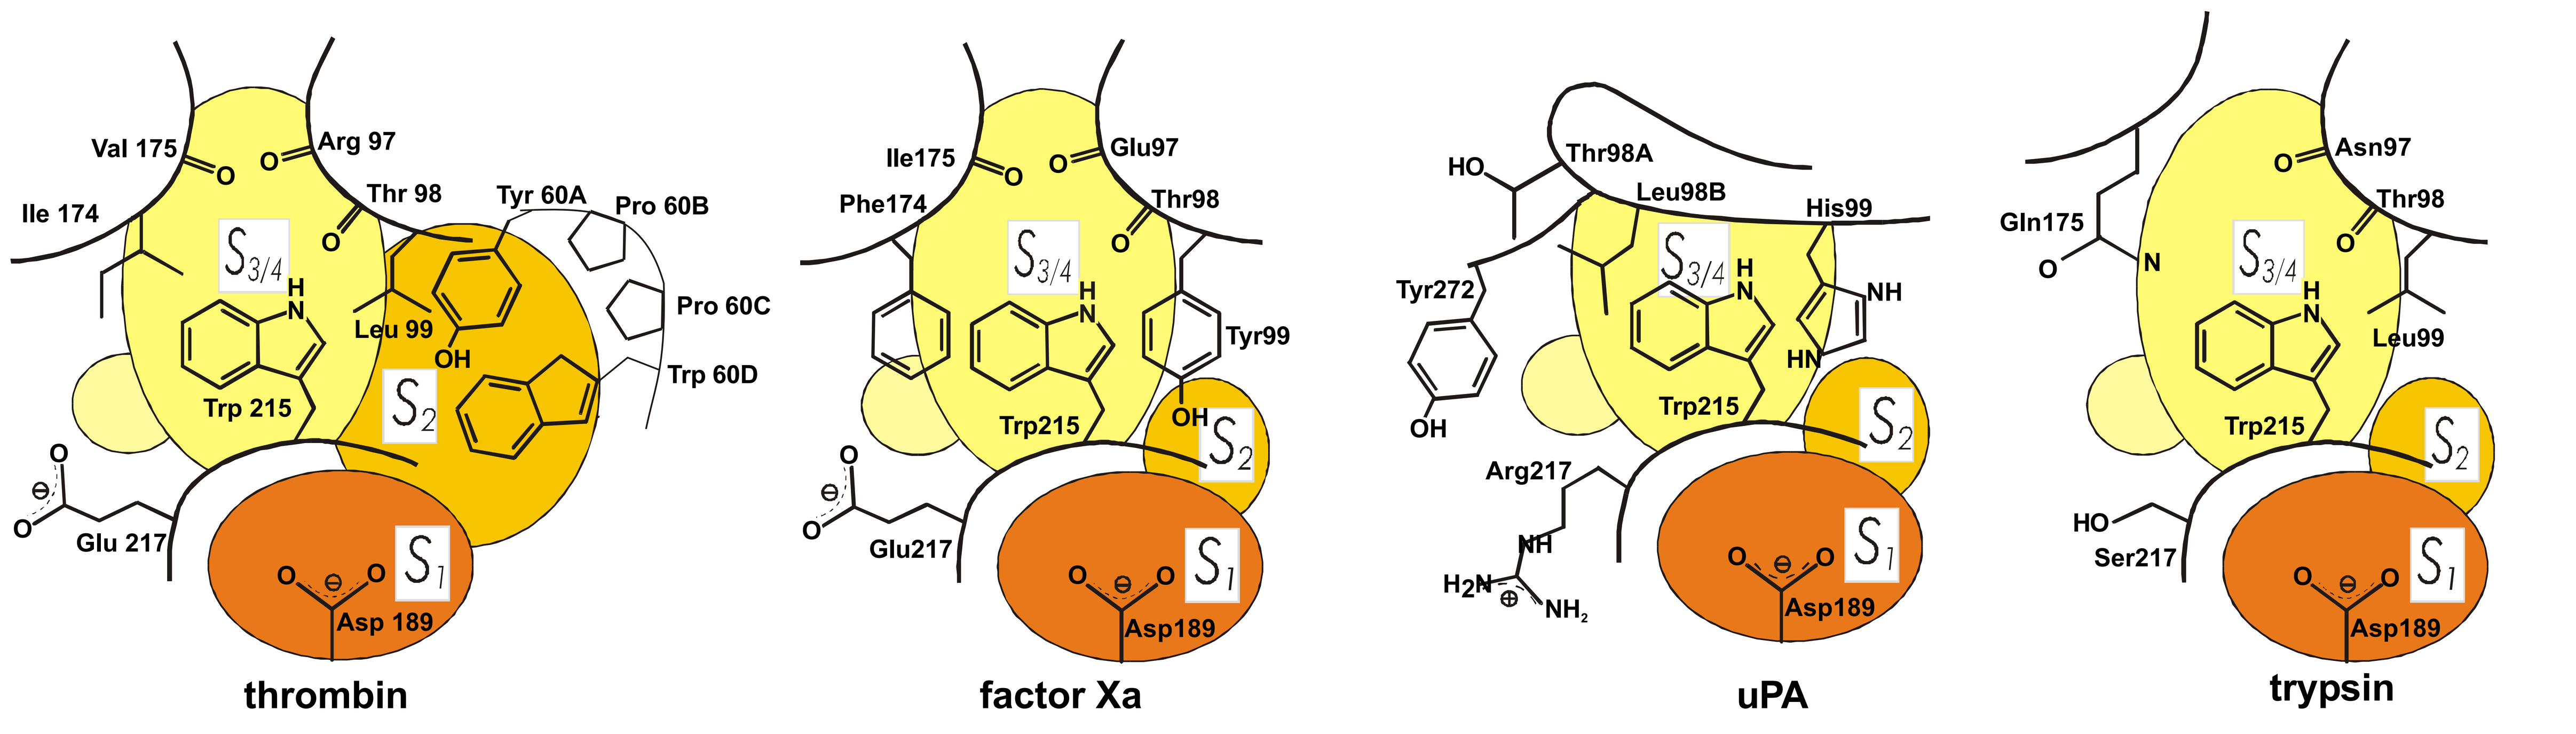
Val 175
Arg 97
Ile 174
Thr 98
Tyr 60A
Pro 60B
Leu 99
Pro 60C
Trp 60D
Trp 215
Glu 217
Asp 189
thrombin
Ile175
Glu97
Phe174
Thr98
Tyr99
Trp215
Glu217
Asp189
factor Xa
Thr98A
Leu98B
His99
Tyr272
Trp215
Arg217
Asp189
uPA
Asn97
Thr98
Gln175
Leu99
Trp215
Ser217
Asp189
trypsin

References

1. Wade, R.C., Henrich, S., Wang, T. (2004) Using 3D protein structures to derive 3D-QSARs. *Drug Discovery Today: Technologies* 1:241-246.
2. Ortiz, A.R., Pisabarro M.T., Gago F., Wade R.C. (1995) Prediction of drug binding affinities by comparative binding energy analysis. *J. Med. Chem.* 38:2681-2691.
3. Wade, R.C., Ortiz, A.R., Gago F. (1998) Comparative Binding Energy Analysis. *Persp. Drug Disc. and Des.* 9:19-34.
4. Wang, T., Wade, R.C. (2001) Comparative Binding Energy (COMBINE) Analysis of Influenza Neuraminidase-Inhibitor Complexes. *J. Med. Chem.* 44:961-971.

Figure 1 (left): The trypsin-like serine proteases of the blood coagulation cascade and those who are related to it because of their side effects. The proteases have structurally very similar binding pockets in the active site.
(Figure was adopted from S. Sperl, http://tumb1.biblio.tu-muenchen.de/publ/diss/ch/2000/sperl.html)